Abstract 1888
Poster F1-1973

Pharmacokinetic / Pharmacodynamic Profile of MK-2764 / PTK 0796 against *S. pneumoniae* in a Murine Pneumonia Model

P. R. TESSIER[1], H. W. FAN[1], S. K. TANAKA[2], D. P. NICOLAU[1]

[1]Ctr. for Anti-Infect Res. & Dev., Hartford Hosp., Hartford, CT USA ~ [2]Paratek Pharmaceuticals, Boston, MA USA

Center for Anti-Infect R & D
Hartford Hospital
80 Seymour Street
PO Box 5037
Hartford, CT 06102-5037
t: 860 . 545 . 3941
f: 860 . 545 . 3992

ABSTRACT

Background A leading causative agent of community-acquired pneumonia (CAP), *Streptococcus pneumoniae* (SPN), has developed resistance to several classes of antimicrobials. MK-2764 / PTK 0796, a novel aminomethylcycline antibiotic derived from the tetracycline class, has demonstrated potent *in vitro* activity against pulmonary pathogens, including multi-drug resistant SPN.

Methods The efficacy of MK-2764 was assessed against penicillin-susceptible (PEN-S) and penicillin/macrolide-resistant (PEN/MAC-R) SPN isolates in a neutropenic mouse pneumonia model. The pharmacokinetic profile of MK-2764 in serum was described at doses of 0.5 to 10 mg/kg after single subcutaneous doses. PK modeling techniques were used to characterize the $_{free}$MK-2764 drug exposure after various single and multiple dose regimens. Evaluation of PD parameters and estimations of the effective dose by bacterial density measurement were completed. Accumulation of MK-2764 into epithelial lining fluid (ELF) and lung tissue was also measured.

Results MK-2764 displayed a linear PK profile over the range of 0.5 to 10 mg/kg doses used in PD studies. Average initial parameters were as follows: V_F (L/kg) 6.99; k01 (1/hr) 38.96; k10 (1/hr) 0.107. Potent efficacy of 2 ½ logs bacterial kill by MK-2764 was achieved after doses of 10 mg/kg/day. For both strains, the static dose was 1 mg/kg. The $_{free}$AUC/MIC was predictive of the change in bacterial load. Concentration (AUC) ratios for ELF:serum and lung tissue:serum ranged from 1.1 to 1.4 and 2.3 to 4.4, respectively.

Conclusions MK-2764 was an effective agent against SPN in this *in vivo* model and the PD parameter predictive of antibacterial activity was the $_{free}$AUC/MIC. Both ELF and lung tissue concentrations indicated accumulation of MK-2764 in these extravascular compartments. The characteristics of MK-2764 including potent antibacterial activity and accumulation at the site of infection make this a candidate for further evaluation.

INTRODUCTION

MK-2764 / PTK 0796 is the first aminomethylcycline to enter clinical development (Figure 1). MK-2764 is currently in Phase 1 clinical development for both oral and intravenous administration. Understanding the pharmacodynamic drivers for efficacy is a key parameter for selection of appropriate human dosing. Since MK-2764 is targeted for development as a therapy for respiratory infections, this study was undertaken to evaluate and/or define the pharmacodynamic parameters of MK-2764 in an experimental mouse pneumonia model after infection with *Streptococcus pneumoniae*.

Structure of MK-2764 / PTK 0796

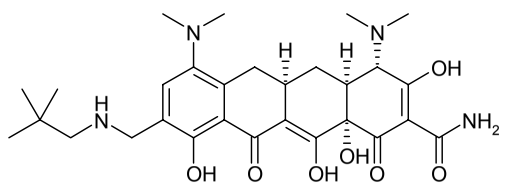

METHODS

Animals Specific-pathogen-free, female ICR (CD-1) mice, weighing between 18-20 grams, were obtained from Harlan Laboratories (Indianapolis, Indiana) and utilized throughout the experiment. Animals were allowed to acclimate approximately one week before experimentation.

Bacteria A penicillin-resistant, *mefA* positive *Streptococcus pneumoniae* (SPN) strain designated #100 and a penicillin-susceptible SPN strain designated #22 were utilized in these *in vivo* experiments.

Antimicrobial Susceptibility Testing The minimal inhibitory concentration (MIC) of MK-2764 was determined for both bacterial isolates by broth microdilution methods performed according to CLSI standards (using 5% lysed horse blood in Cation-adjusted Mueller Hinton broth).

Antibiotic compounds MK-2764 was supplied by Paratek Pharmaceuticals. Both *in vitro* and *in vivo* solutions were prepared in sterile water.

Pharmacokinetic studies Neutropenic ICR mice were prepared as described below for the lung infection model and infected with one of the SPN strains. MK-2764 was administered via subcutaneous injections to groups of 6 mice approximately 12 hours after infection. Blood, bronchial alveolar lavage (BAL) samples and whole lung tissue samples were collected from the same animals from each regimen. The BAL samples that were obtained simultaneous to serum were assayed for urea to allow for the determination of drug levels in epithelial lining fluid (ELF). Analysis for the concentrations of MK-2764 in murine serum, tissue, and BAL was done by LC/MS/MS.

Efficacy Determination of MK-2764 in the murine pneumonia model To establish neutropenia, cyclophosphamide for injection (Cytoxan®, Bristol-Myers Squibb, Princeton, NJ) was administered on the fourth day prior and on the day before infection. The initial dose was 150 mg/kg, the second dose was 100 mg/kg. Administration of the solutions was performed by IP injection in 0.2 ml volume per mouse.

A total of 6 mice per treatment regimen group were infected with a strain of SPN. Pneumonia was induced by instilling 0.05ml of 10^8 CFU/ml SPN suspended in 5% dextrose in normal saline into the lungs by inhalation. Dosing commenced 12-14 hours after lung inoculation (i.e. study 0h) via subcutaneous injection of freshly prepared solutions.

Bacterial cultures of lung tissue were prepared from five lobes of the lungs from each animal. Samples were homogenized, diluted in saline and plated onto agar media plates (Trypticase soy agar with 5% sheep blood) for bacterial growth.

RESULTS

***In vitro* susceptibility of the SPN strains** The MICs of the SPN isolates to the MK-2764 are listed on Table 1. MK-2764 exhibited exceptional activity against both strains demonstrated by the low MIC values.

Table 1: Susceptibility characteristics of the S. pneumoniae isolates

SPN strain	MK-2764 (mg/L)	Penicillin interpretation[a]	Genotypic variations or known resistance
22	0.03	S	None
100	0.06	R	*mefA*

[a]Susceptibility interpretations according to Clinical and Laboratory Standards Institute[1]: S = susceptible; I = intermediate; R = resistant

Pharmacokinetics of MK-2764 in SPN-infected mice Individual pharmacokinetic parameters for the 0.5, 5, 10, 50 and 100 mg/kg doses are recorded on Table 2.

Table 2: Pharmacokinetics of MK-2764 after subcutaneous dosing regimens in SPN-infected mice

Dosing Regimen (mg/kg)	Calculated Cmax (mg/L)	Tmax (hr)	AUC_{0-24} (mg*hr/L)	V_F (L/kg)	k01 (1/hr)	k10_HL (hr)	Cl (ml/hr/kg)
0.5	0.063	0.153	0.56	7.86	38.91	6.77	0.80
5	0.542	0.119	5.07	9.12	52.96	7.00	0.90
10	2.45	0.215	18.54	3.98	25.00	5.84	0.47
50	6.99	1.213	N/A	N/A	N/A	N/A	N/A
100	11.64	1.659	N/A	N/A	N/A	N/A	N/A

The C_{max} and AUC values were found to be linear between the 0.5 mg/kg and 10 mg/kg doses. Dosing regimens used in the efficacy studies were simulated based on the following average initial parameters: V_F (L/kg) 6.99; k01 (1/hr) 38.96; k10 (1/hr) 0.107. Pharmacokinetic parameters for calculations of PD were adjusted to account for free drug fractions.

The ratios of MK-2764 concentration as measured by AUC over 24 hours in mice in epithelial lining fluid and lung tissue versus serum are listed on Table 3. MK-2764 ELF concentrations were slightly greater than in plasma. MK-2764 accumulated in lung tissue to approximately twice that in the ELF.

Table 3: The AUC_{24} ratio of MK-2764 in epithelial lining fluid (ELF) (mg/L) or lung tissue (mg/g) and serum

MK-2764 Dose	ELF: Serum	Lung Tissue: Serum	Lung Tissue: ELF
0.5 mg/kg	1.12	3.08	2.76
5 mg/kg	1.19	4.35	3.64
10 mg/kg	1.39	2.27	1.63

Efficacy of MK-2764 against SPN strains 100 and 22 A dose-dependent response was noted over the range of MK-2764 doses between 0.25 and 10 mg/kg or 0.25 and 50 mg/kg against SPN 22 or 100, respectively, with as much as -2.7 log change at the high doses. (Figure 1 and Table 4) The MK-2764 dose of 1 mg/kg resulted in a net static effect against both SPN 100 and 22. A high correlation (r^2 = 0.904, 0.699) was identified between the AUC/MIC and log_{10} CFU/ml against both SPN 22 and SPN 100.

Figure 1: Change in SPN 22 log_{10} CFU for MK-2764 after subcutaneous dosing regimens versus initial bacterial density

A. Correlation to AUC

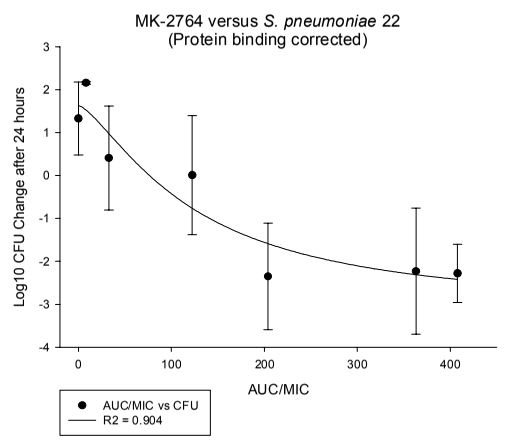
MK-2764 versus S. pneumoniae 22
(Protein binding corrected)
Log10 CFU Change after 24 hours
AUC/MIC
AUC/MIC vs CFU
R2 = 0.904

B. Correlation to Cmax

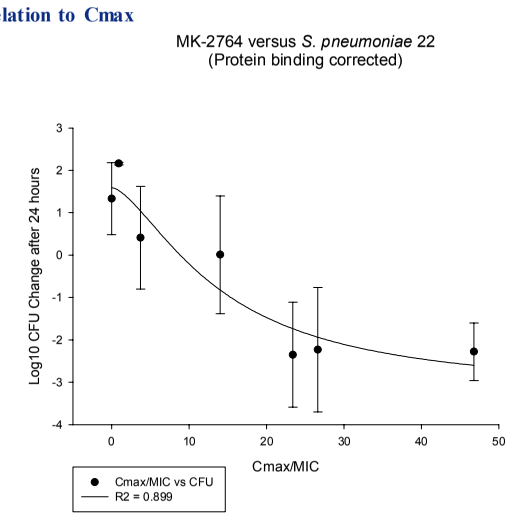
MK-2764 versus S. pneumoniae 22
(Protein binding corrected)
Log10 CFU Change after 24 hours
Cmax/MIC
Cmax/MIC vs CFU
R2 = 0.899

C. Correlation to %Time > MIC

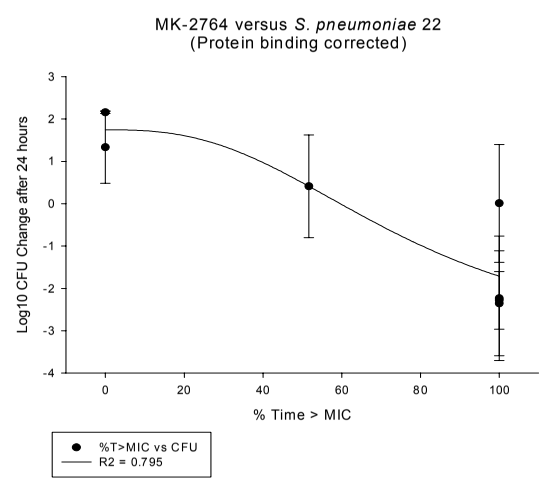
MK-2764 versus S. pneumoniae 22
(Protein binding corrected)
Log10 CFU Change after 24 hours
% Time > MIC
%T>MIC vs CFU
R2 = 0.795

Table 4: Correlation of PK to Efficacy

SPN Test Strain	Correlation (R^2) to: AUC	Time	CMax
22	0.904	0.795	0.899
100	0.699	0.825	0.616

Graphs of the fractionated total daily doses of 3 and 8 mg/kg/day versus the change in log_{10} CFU/ml of SPN 100 after 24 hours are illustrated in Figure 2. A significant reduction in CFU/ml with increasing exposure as measured by AUC/MIC whereas a relationship to C_{max}/MIC or %Time>MIC was not readily apparent. As a result it appears from the dose fractionation studies and evaluation of the composite curve exposures used to evaluate the magnitude of the pharmacodynamic parameters relative to reduction in CFU that the AUC/MIC is the parameter that best expresses the relationship between a PD parameter and efficacy.

Figure 2: Dose fractionation effects on efficacy

A. Correlation to AUC

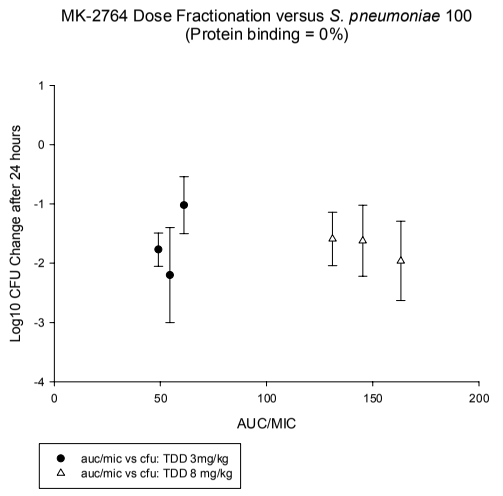
MK-2764 Dose Fractionation versus S. pneumoniae 100
(Protein binding = 0%)
Log10 CFU Change after 24 hours
AUC/MIC
auc/mic vs cfu: TDD 3mg/kg
auc/mic vs cfu: TDD 8 mg/kg

B. Correlation to Cmax

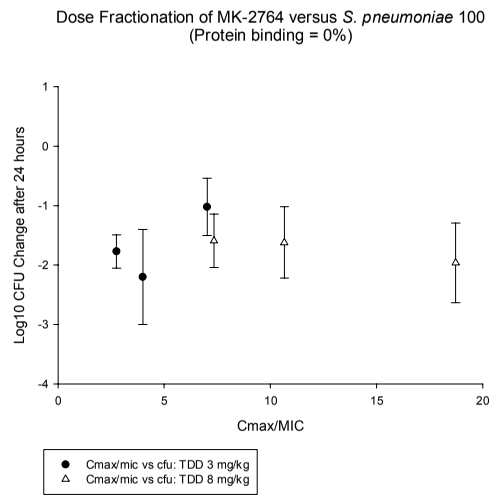
Dose Fractionation of MK-2764 versus S. pneumoniae 100
(Protein binding = 0%)
Log10 CFU Change after 24 hours
Cmax/MIC
Cmax/mic vs cfu: TDD 3 mg/kg
Cmax/mic vs cfu: TDD 8 mg/kg

C. Correlation to %Time > MIC

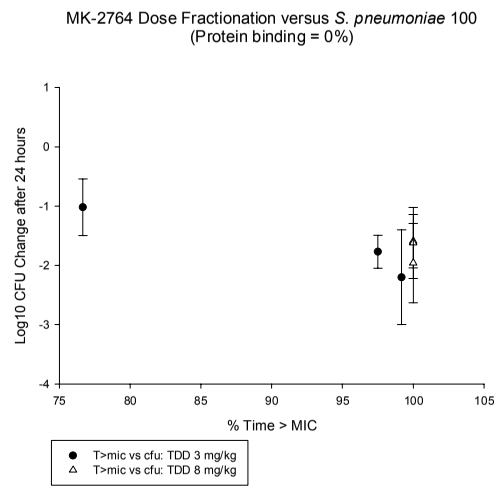
MK-2764 Dose Fractionation versus S. pneumoniae 100
(Protein binding = 0%)
Log10 CFU Change after 24 hours
% Time > MIC
T>mic vs cfu: TDD 3 mg/kg
T>mic vs cfu: TDD 8 mg/kg

CONCLUSIONS

- MK-2764 exhibited activity both *in vitro* and *in vivo* against the two SPN isolates. The presence on *mefA* genotype and penicillin resistance in SPN 100 did not appear to limit or alter the activity of MK-2764 whether *in vitro* or *in vivo* versus SPN 22 which is an antibiotic susceptible strain.
- The pharmacokinetics after single subcutaneous MK-2764 doses of 0.5, 5 and 10 mg/kg were linear and were satisfactorily exemplified using a nonlinear one compartment model.
- MK-2764 appears to concentrate at the site of infection in this model as demonstrated by the amount of drug entry and accumulation in the extravascular compartment of relevance.
- MK-2764 produced an overall dose-dependent killing effect. On an exposure basis, the bacterial density decreased proportionally with an increase of AUC/MIC and a potential impact of C_{max}/MIC. This relationship was found to be favorably correlated for both SPN isolates. The maximum bacterial kill was observed after doses of approximately 10 mg/kg and approached bactericidal activity (i.e. 2.4-2.7 logs).